Evaluation of the Effect of GliSODin®
on the Intensity of Actinic Erythema
Induced with Radiation

S. Mac-Mary[1], J.M. Sainthillier[1], P. Creidi[1], J.P. Series[2],
F. Vix[2], Ph. Humbert[1].

[1]**Laboratoire d'Ingénierie et de Biologie Cutanées (LIBC),**
IFR Ingénierie et biologie cellulaire et tissulaire
Service de Dermatologie, Centre Hospitalo Universitaire
25030 BESANCON Cedex, FRANCE

[2]**Laboratoire Isocell,**
Bd du Général Valin,
Paris, France

Introduction

GliSODin® has already demonstrated activation properties on the principal internal antioxidants, in particular Superoxide Dismutase (SOD), Catalase and Glutathione Peroxidase. The purpose of this trial was to assess its efficacy in solar prevention and protection.

Materials and methods

- 50 healthy subjects (10 phototype II, 20 phytotype III and 20 phototype IV)
- Evaluation of the MED (Minimum Erythematous Dose) before and after 4 weeks of the daily administration of GliSODin or a placebo (Day 1 and Day 30)
- Monitoring of the actinic erythematous induced on the forearm (inside) of these subjects by chromametry (redness, a*) 24 hours after irradiation exposure (3 x MED) and then every week for 4 weeks (W0, W1, W2, W3, W4).
- Measurements completed by an analysis of the capillary network by videocapillaroscopy.

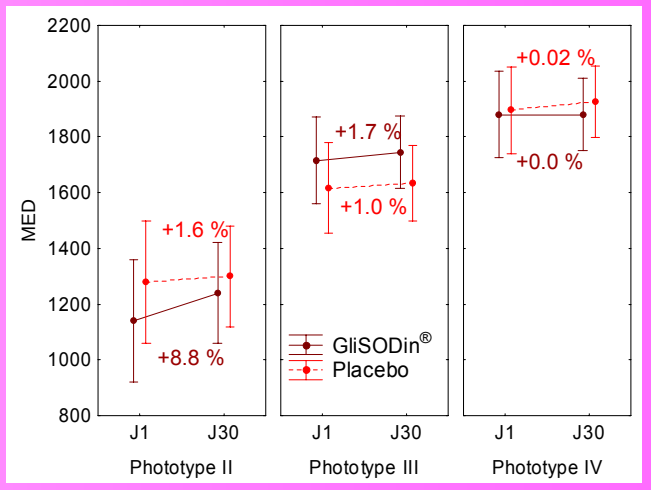
2200
2000
1800
1600
1400
1200
1000
800
MED
+1.6 %
+8.8 %
+1.7 %
+1.0 %
+0.02 %
+0.0 %
GliSODin®
Placebo
J1
J30
Phototype II
Phototype III
Phototype IV

Fig 1. Evaluation of the MED as a function of time, phototype and treatment

Results

- Increase in the MED in the GliSODin group, particularly in the light phototypes (II and III) (Figure 1)
- Faster reduction in redness with Glisodin® than with placebo (Figure 2).
- Faster increase in the capillary density than with the placebo (Figure 2).

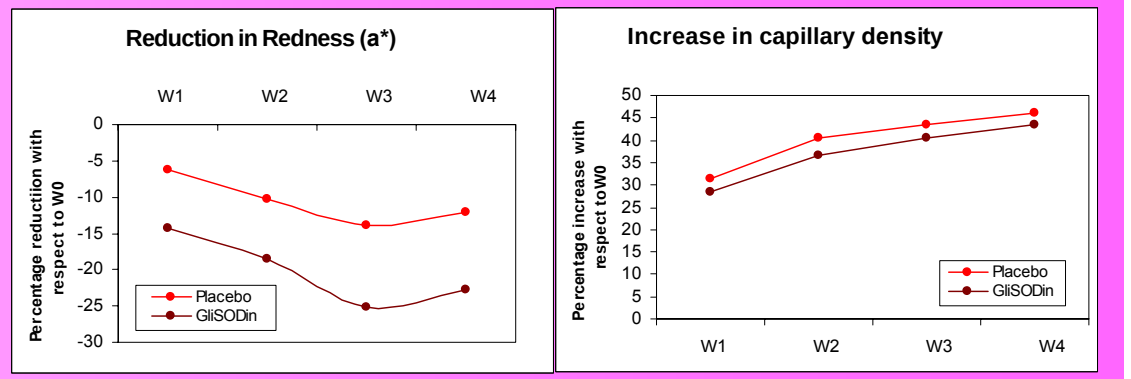
Reduction in Redness (a*)
W1
W2
W3
W4
0
-5
-10
-15
-20
-25
-30
Percentage reductiion with respect to W0
Placebo
GliSODin
Increase in capillary density
50
45
40
35
30
25
20
15
10
5
0
Percentage increase with respect toW0
Placebo
GliSODin
W1
W2
W3
W4

Fig 2. Evolution of redness and skin microcirculation as a function of time and treatment

Discussion

GliSODin® protects the cells against the negative effects of oxidative stress [1] by remaining active during intestinal passage [2], by activating the internal system of antioxidant enzyme defenses [3] and by limiting cell death resulting from oxidative stress. [4]. All of the results in this trial indicate the action of GliSODin® in solar protection. It would be interesting to test the efficacy of GliSODin® in this type of indication as a pre-treatment and to repeat the trial on a larger number of subjects, in particular on the light phototypes (II).

Références

[1] MUTH CM, GLENZ Y, KLAUS M, et al. *Free Rad Res* 2004 ; **38** : 927-932.
[2] VOULDOUKIS I, LACAN D, KAMATE C, et al. *J Ethnopharmacol* 2004 ; **94** : 67-75.
[3] VOULDOUKIS I, CONTI M, KOLB JP, et al. *Res Trends – Curr Trends Immunol* 2003 ; **5** : 151-145.
[4] VOULDOUKIS I, CONTI M, KRAUSS P, et al. *Phytoterapy Res* 2004 ; **18** : 957-962.